1nHealth
WE OWN ENROLLMENT

Challenging The AI Default

How human insights and operational excellence drove 250% more enrollments than expected in a psychedelic postpartum depression trial.

Dan Brenner, Kayt Leonard, Steve Wimmer, Alyssa Beck

Objective

To evaluate whether a patient-centric recruitment model, anchored in human-led prescreening, site engagement, and real-time data science, could accelerate enrollment in a postpartum depression (PPD) psychedelic treatment study.

Design

The sponsor initially engaged 1nHealth to contribute 25% of total trial enrollment, with particular emphasis on maintaining candidate quality throughout the recruitment process.

From the outset, we applied our core operating model: real humans guiding patients through prescreening conversations, close collaboration with sites to align expectations, and continuous tracking of every stage in the enrollment funnel, from submission to screening, ICF, and randomization.

This hands-on, data-informed approach allowed us to identify bottlenecks early, refine messaging to improve conversion rates, and ensure that each candidate advanced through the process efficiently and appropriately. Because our infrastructure supports rapid creative testing and immediate operational pivots based on real-time performance data, we were able to maintain both velocity and precision in patient recruitment. The result was not only meeting our enrollment targets but doing so with a consistent focus on quality, transparency, and partnership with both sponsors and sites.

Human Insights: Resonance Drives Quality

Through iterative A/B testing, we identified visual styles that both engaged and aligned with the psychedelic theme of evocative gradients, organic motion, and softened imagery that mirrored internal transformation, driving stronger connection and click-through rates.

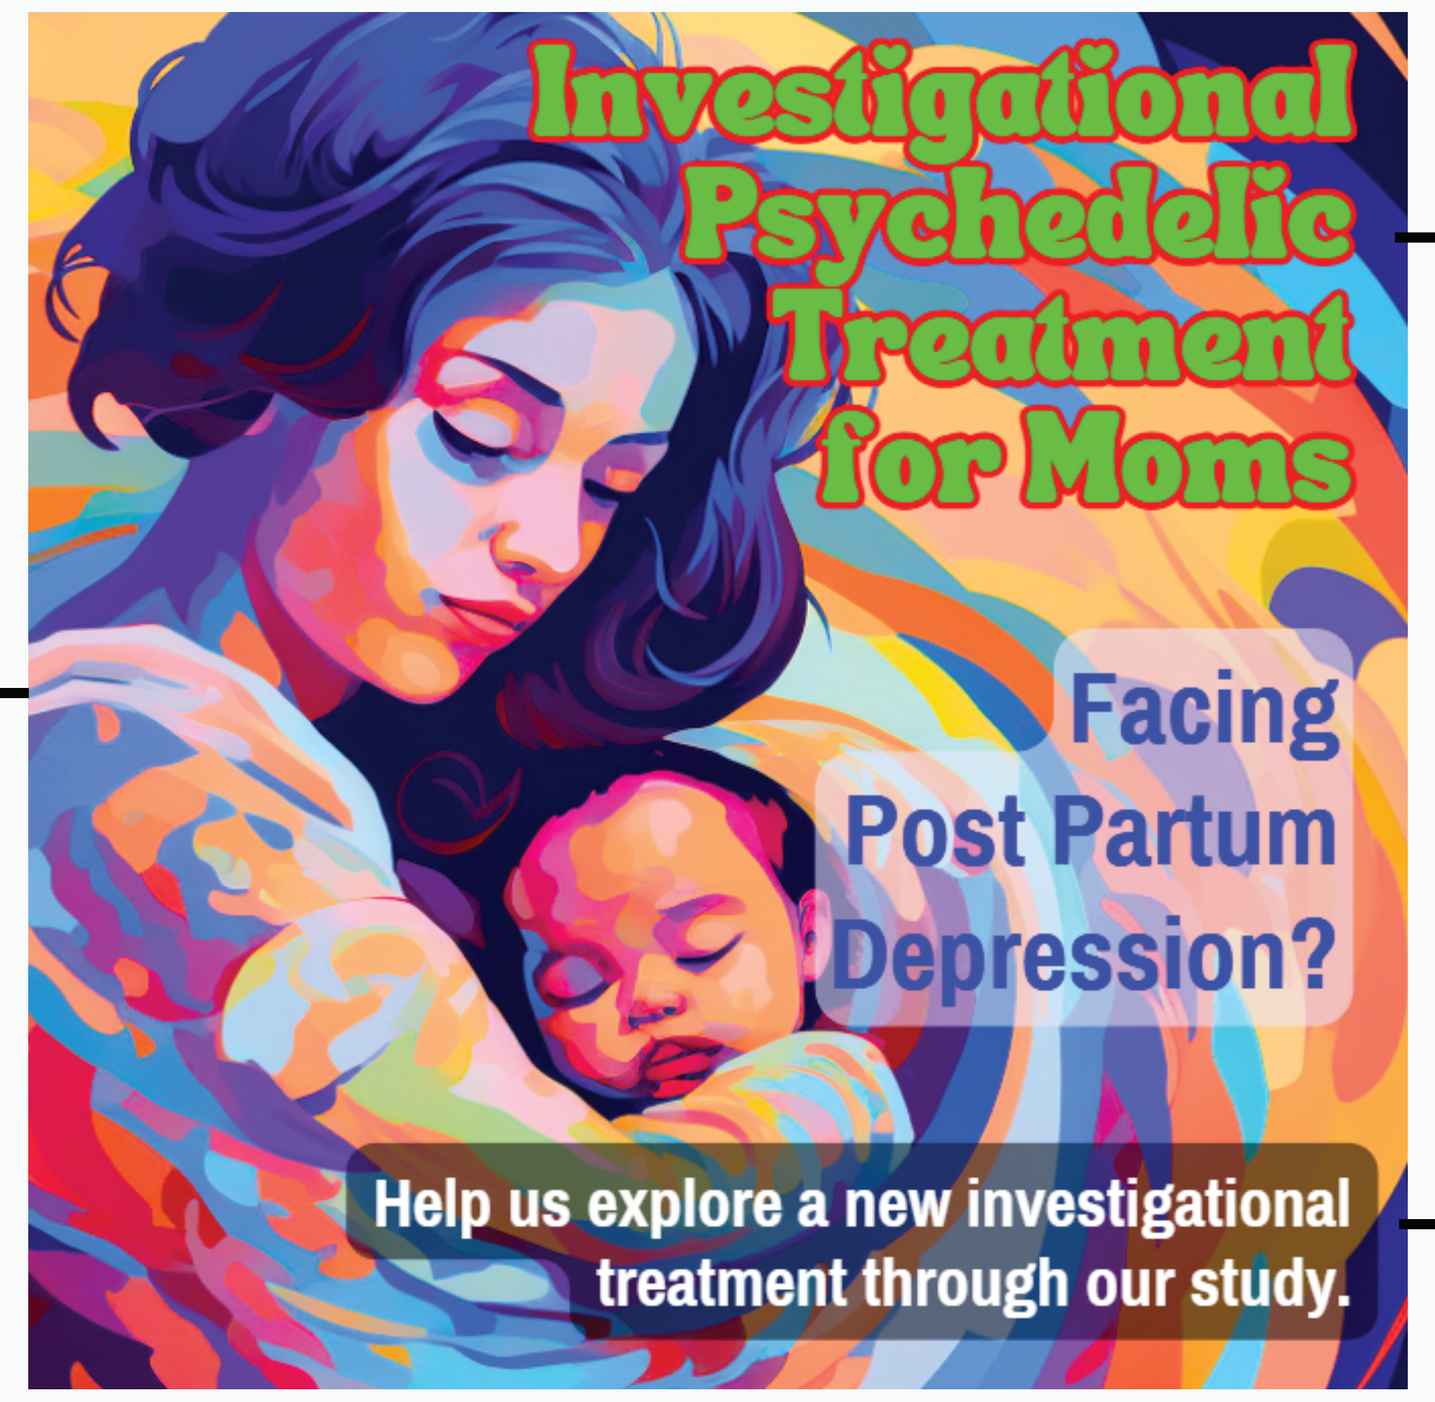
Investigational Psychedelic Treatment for Moms
Facing Post Partum Depression?
Help us explore a new investigational treatment through our study.

Early qualitative insights showed that patients valued transparency. By naming the treatment as psychedelic early, we built trust and curiosity rather than hesitation.

Calling it an investigational treatment reframed "experimental" as "a new chance" for women who had exhausted other options and were looking for hope.

Operational Excellence: Tailored For New Moms

Empathy-Led Coordination
Only female patient coordinators interacted with new moms, creating an atmosphere of trust and safety.

Follow-Through That Cares
More follow-ups acknowledged real-life unpredictability, re-engaging participants after missed visits.

Gentle, Frequent Touchpoints
Increased appointment reminders and nurturing communications reduced stress and supported adherence.

Flexibility As Compassion
Offering flexible scheduling options honored the realities of early motherhood and kept participation attainable.

Results

Month-To-Month Enrollment

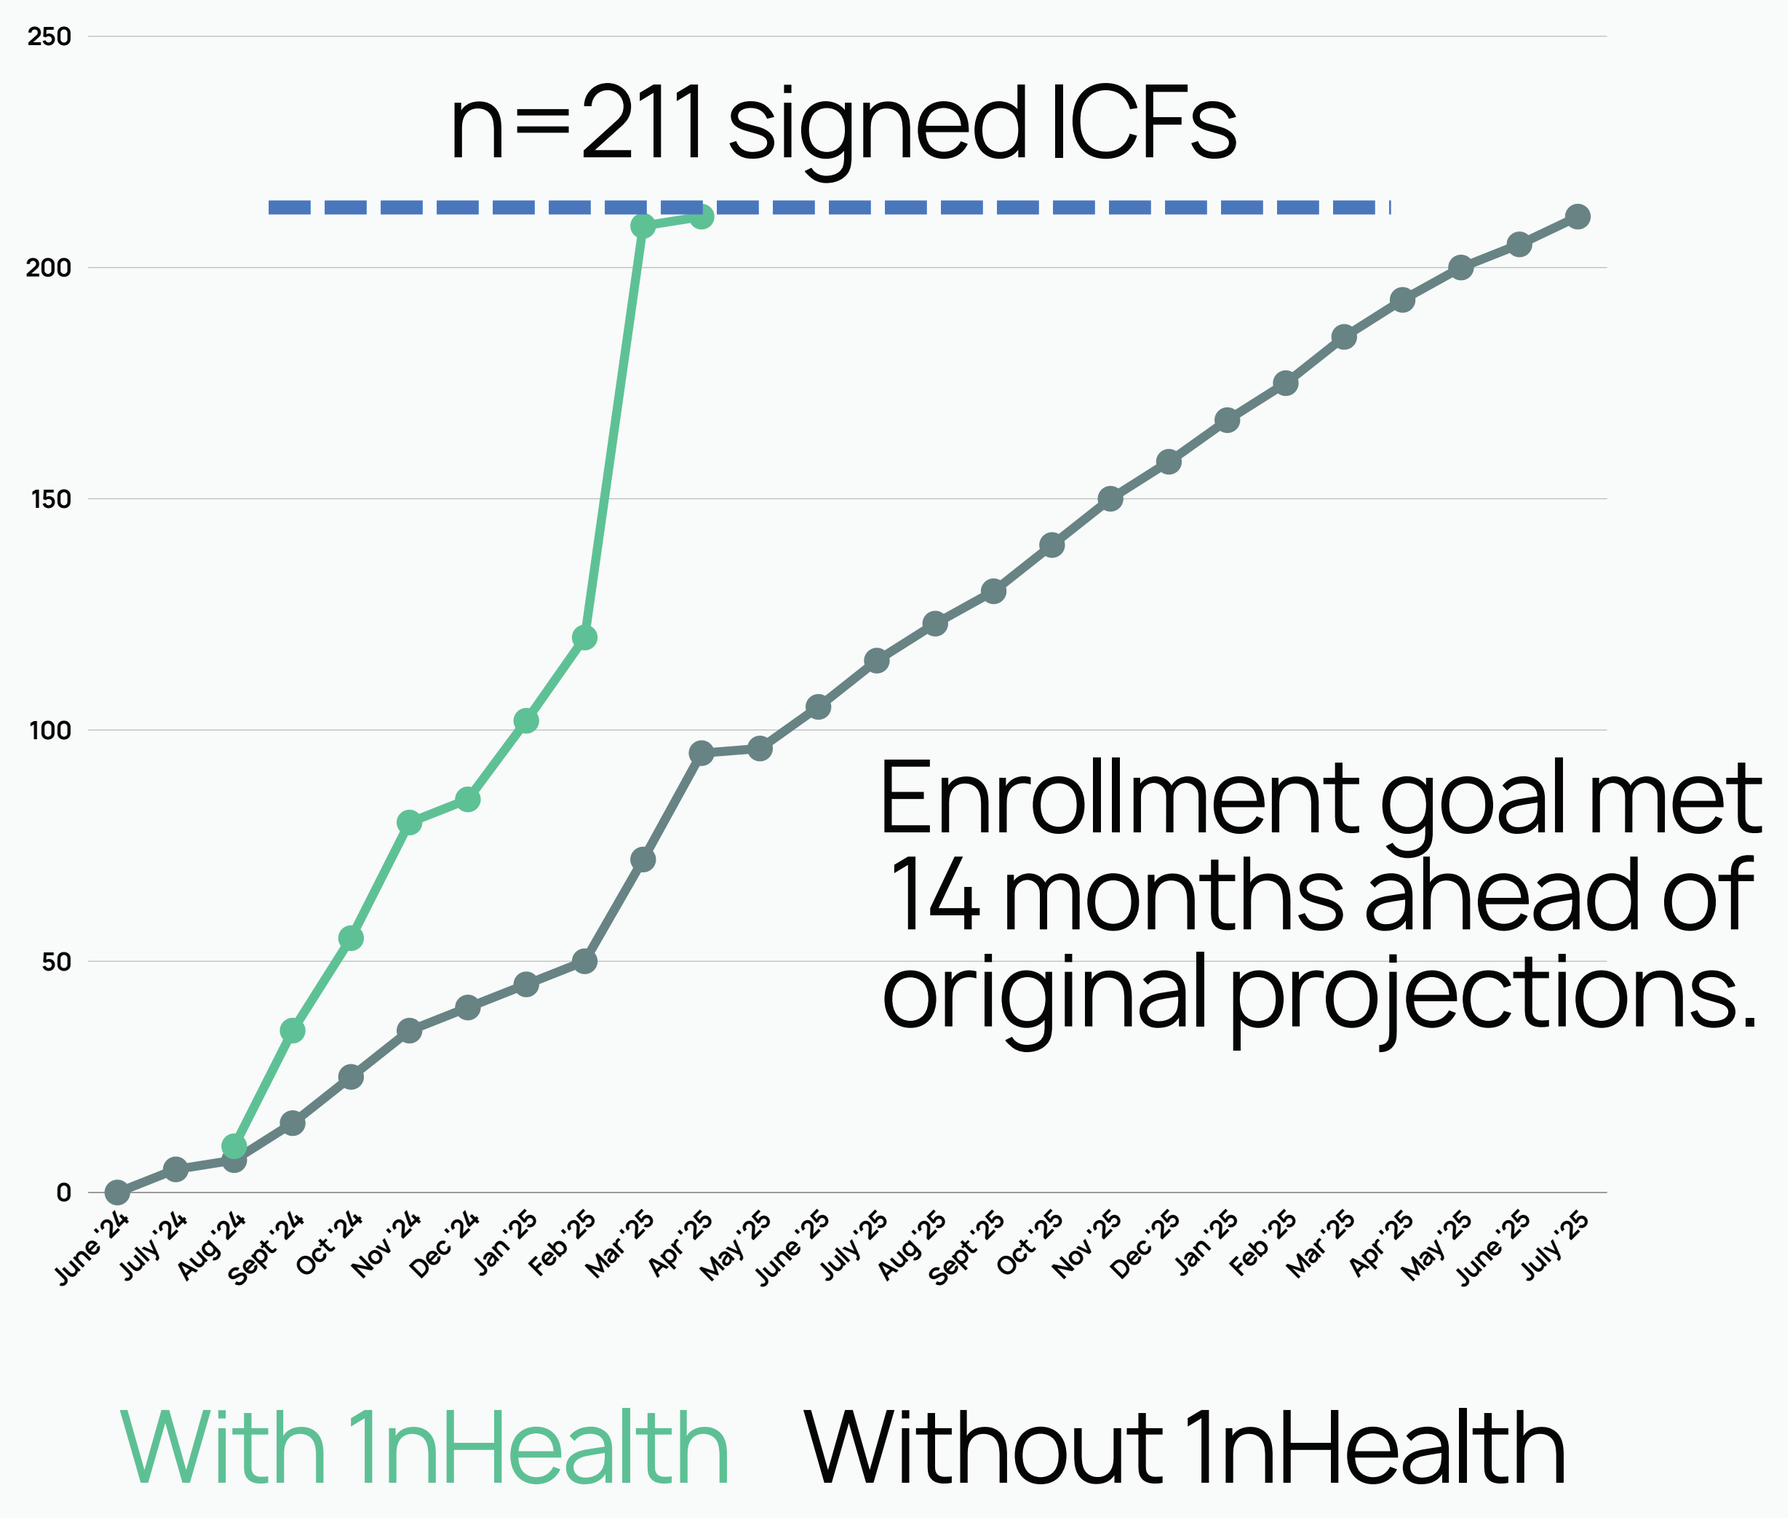
n=211 signed ICFs
Enrollment goal met 14 months ahead of original projections.
250
200
150
100
50
0
June '24
July '24
Aug '24
Sept '24
Oct '24
Nov '24
Dec '24
Jan '25
Feb '25
Mar '25
Apr '25
May '25
June '25
July '25
Aug '25
Sept '25
Oct '25
Nov '25
Dec '25
Jan '25
Feb '25
Mar '25
Apr '25
May '25
June '25
July '25
With 1nHealth
Without 1nHealth

Majority of consented patients (54%)

Majority of randomized patients (56%)

Lowest screen fail rate (58%)

Conclusion

Recruitment success was driven not by bold creative alone, but by operational excellence such as the disciplined execution of human-led patient engagement, real-time analytics, and a culture of rapid testing and adaptation.

This integrated approach ensured that every decision was informed by data and grounded in collaboration. The result was a higher proportion of qualified patients, stronger site relationships built on trust and transparency, and enrollment outcomes that surpassed sponsor expectations in both speed and quality.

Read more about this study and others by scanning the QR code: