1nHealth
WE OWN ENROLLMENT

Adaptive Recruitment In Action

Delivering on time when traditional enrollment methods fail.

Dan Brenner, Kayt Leonard, Steve Wimmer, Alyssa Beck

Objective

This case study demonstrates how proactive and flexible recruitment strategies can meet enrollment goals under manufacturing-driven timelines. In a chronic fecal incontinence gene therapy trial, the study team faced immovable operational deadlines and a limited recruitment window. Through adaptive planning, real-time decision-making, and rapid execution, 1nHealth maintained both the speed required for delivery and the quality necessary for trial integrity. The result illustrates how strategic agility and close sponsor collaboration can turn fixed constraints into success.

Design

The study faced a hard manufacturing cutoff, creating significant time pressure to complete enrollment. Enrollment model validation confirmed that traditional recruitment methods would fall short of the required pace, prompting 1nHealth to design and deploy a rapid-response strategy.

A pilot campaign was launched immediately to begin patient recruitment, with infrastructure in place to scale seamlessly into a full campaign within months.Throughout the process, transparent communication and close c ollaboration with the sponsor were essential, allowing rapid alignment on unconventional, data-backed tactics that ultimately proved efficient and effective.

Customizing Enrollment Under Pressure

Feasibility First: Modeled recruitment timelines for accuracy.

Pre-Tested Messaging: A/B tested creative pre-launch.

Sequential Site Start: Activated sites one by one.

Verified via EDC: Reconciled site data post-visit.

Refine, Then Scale: Optimized before full roll-out.

Standard Enrollment Campaign

Pilot Launch: Began recruiting while building campaign.

Live Optimization: A/B tested creative in real time.

Early Site Buy-In: Engaged sites before activation.

Joint EDC Review: Reconciled data with sponsor.

Scale in Motion: Expanded as results confirmed.

Accelerated Enrollment Campaign

Results

Month-To-Month Consents

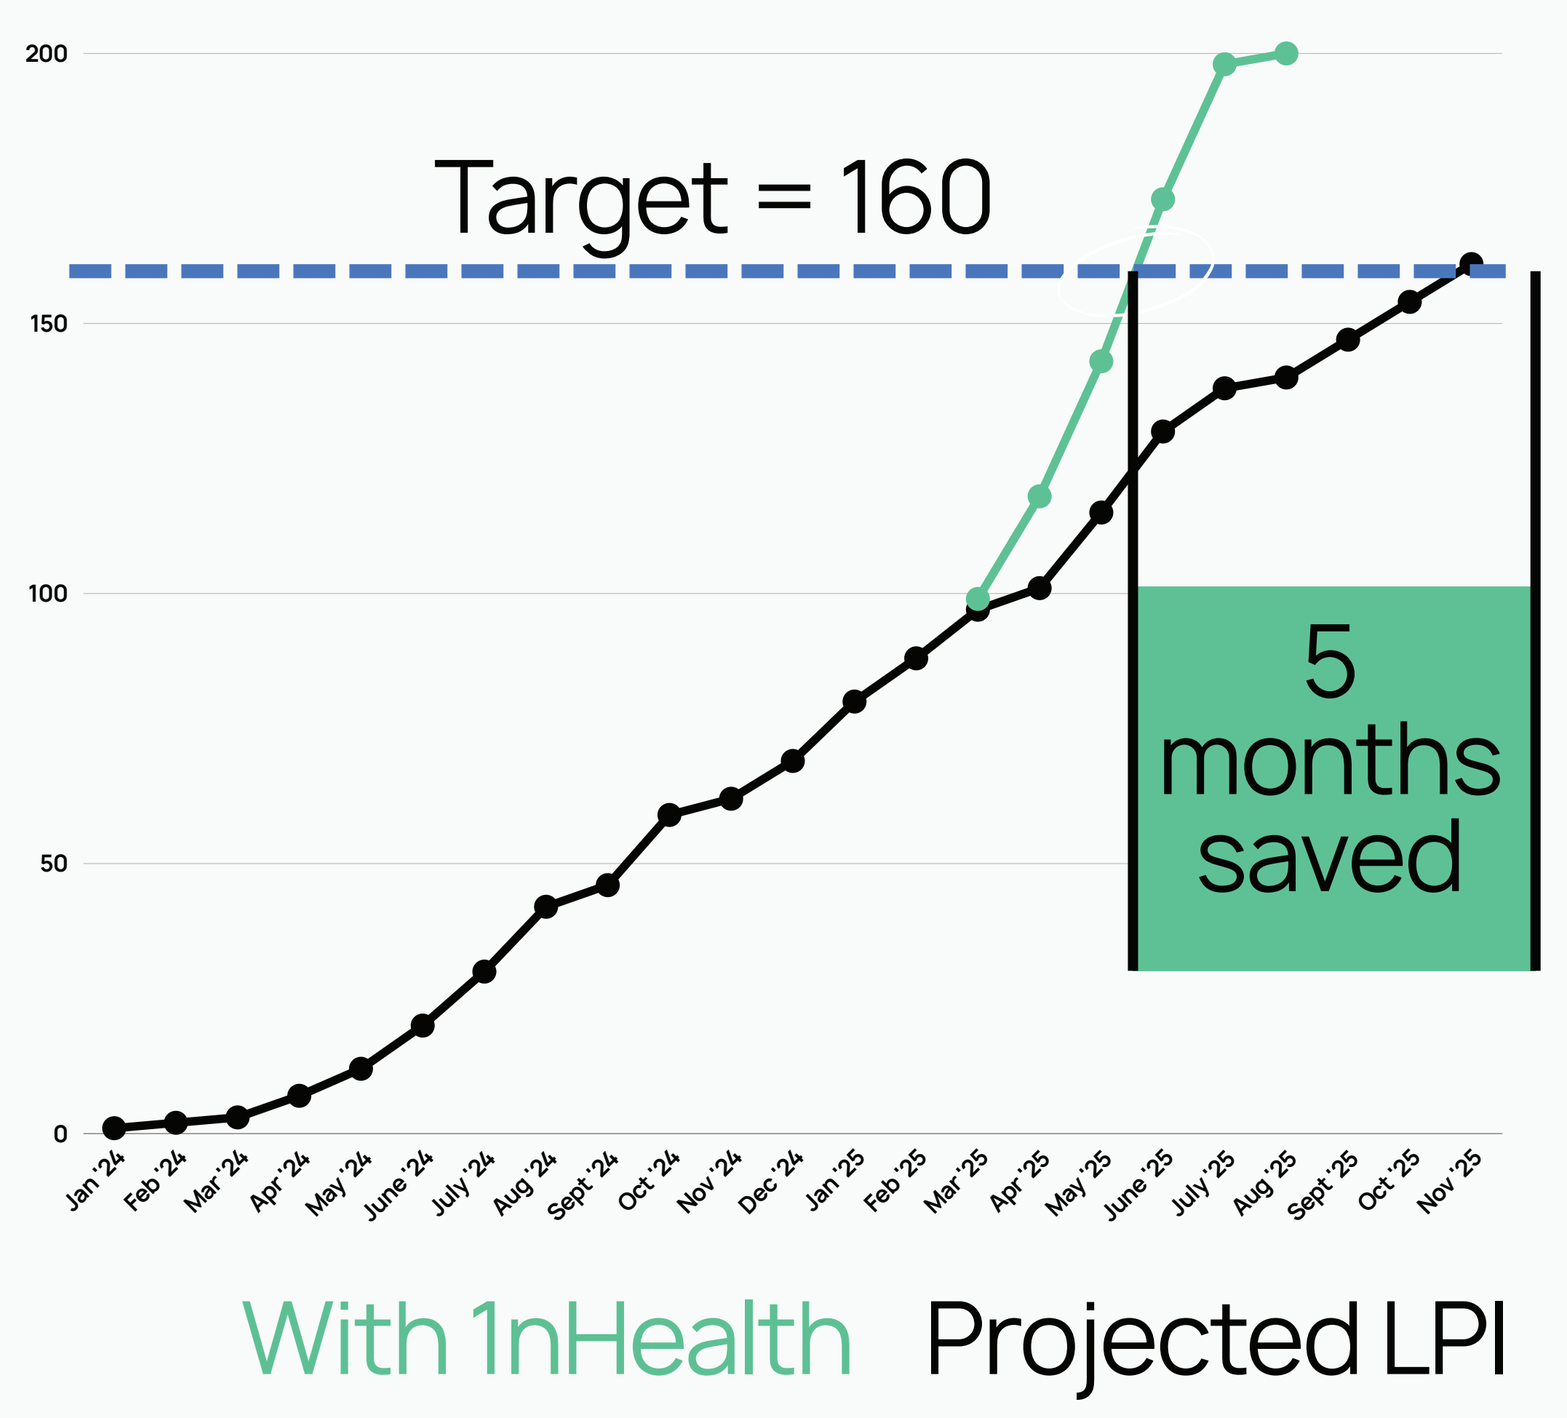
Target = 160
200
150
100
50
0
Jan '24
Feb '24
Mar '24
Apr '24
May '24
June '24
July '24
Aug '24
Sept '24
Oct '24
Nov '24
Dec '24
Jan '25
Feb '25
Mar '25
Apr '25
May '25
June '25
July '25
Aug '25
Sept '25
Oct '25
Nov '25
5 months saved
With 1nHealth
Projected LPI

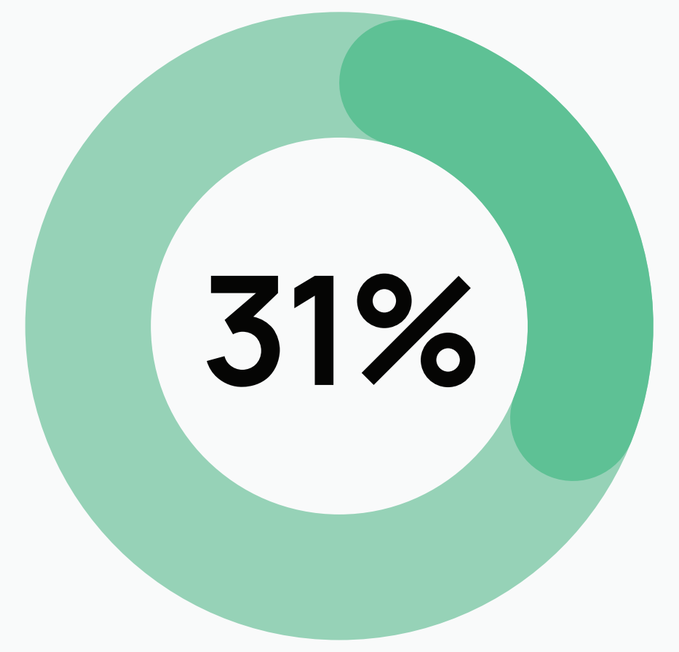
31%

of all randomizations sourced by 1nHealth.

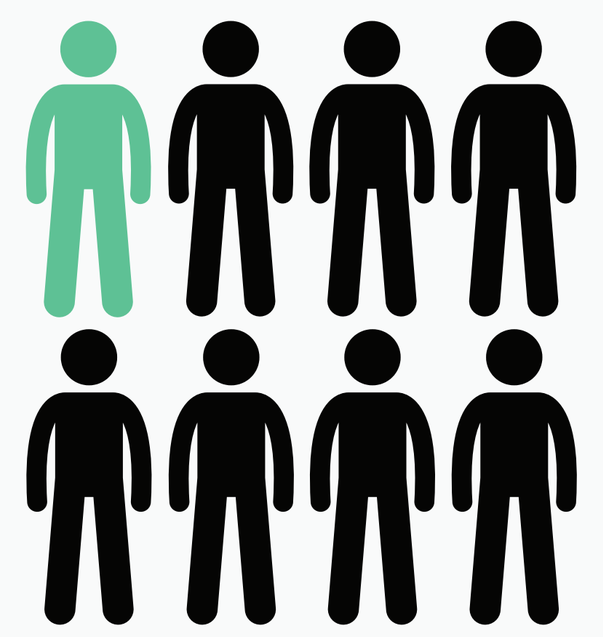

1 out of 8 patients who passed phone screening were randomized.

Conclusion

When traditional recruitment models can't keep pace with accelerated timelines, proactive piloting and flexible, data-driven strategies become essential to mitigating risk and protecting study deliverables.

That approach not only overcame the challenges of a fixed manufacturing deadline, but also demonstrated how adaptability, rapid decision-making, and strong sponsor–partner collaboration can sustain trial momentum under intense operational pressure. The outcome reinforces that agility is not just an advantage in clinical research, it's a critical capability.

Read more about this study and others by scanning the QR code: